6229

Randomized Controlled Trial: Lumbar Medial Branch Cryoneurolysis Versus Radiofrequency Ablation for Chronic Low Back Pain

Martin G. Ferrillo, Kasandra Cliff

Albany & Saratoga Centers for Pain Management, Saratoga Springs, NY

OBJECTIVE

The objective of this pilot study was to compare the effect of lumbar medial branch cryoneurolysis to radiofrequency ablation (RFA) for treatment of chronic low back pain

CONCLUSIONS

1. Cryoneurolysis was well tolerated and associated with durable pain reduction that was significantly superior to RFA 1 year after treatment
2. Cryoneurolysis also led to improvements in functional disability and patients' overall impression of change compared with RFA 1 year after treatment for low back pain

PRESENTING AUTHOR: Martin G. Ferrillo; martingf@aol.com

DISCLOSURES: MGF has received consulting fees from Boston Scientific and Pacira BioSciences, Inc. KC has no relevant financial relationships or support to disclose.

ACKNOWLEDGMENTS: This study was funded by Pacira BioSciences, Inc. Assistance with poster preparation was provided under the authors' direction by Emma Hinkle, PhD, of MedThink SciCom and funded by Pacira BioSciences, Inc.

REFERENCES: **1.** Wray JK, Dixon B, Przkora R. *StatPearls.* Treasure Island, FL: StatPearls Publishing; 2023. **2.** Airaksinen O, Brox JI, Cedraschi C, et al. *Eur Spine J.* 2006;15(suppl 2):S192-S300. **3.** Guirguis M, Hakkinen I, Amiri Y, III JP, Kim CH. *Deer's Treatment of Pain.* Springer; 2019.

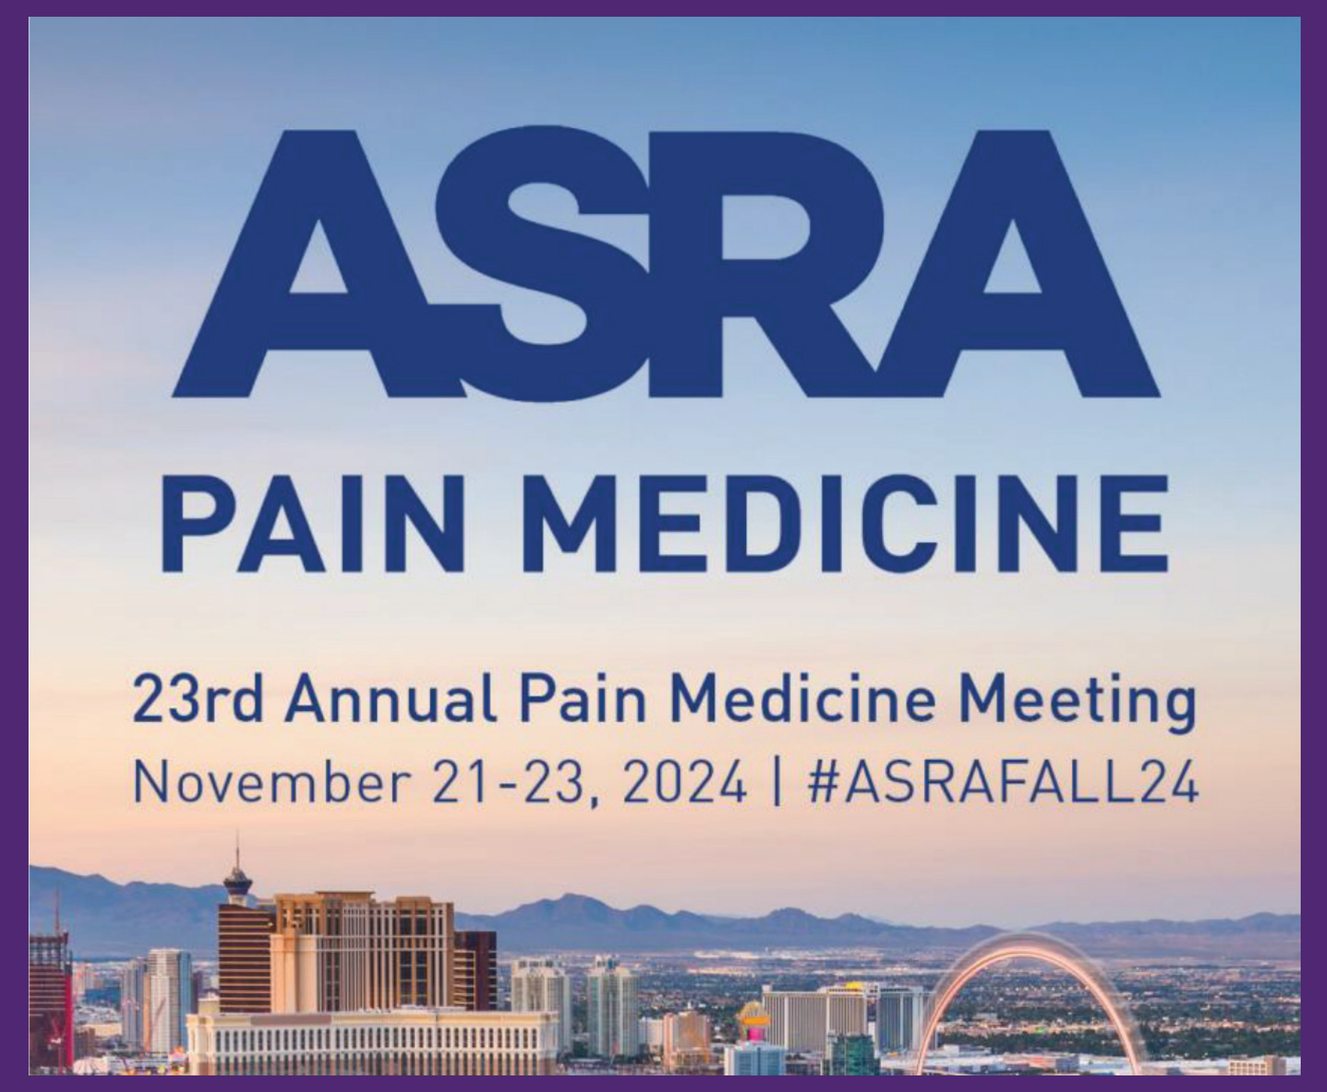

ASRA
PAIN MEDICINE
23rd Annual Pain Medicine Meeting
November 21-23, 2024 | #ASRAFALL24

Presented at the American Society of Regional Anesthesia and Pain Medicine Annual Pain Medicine Meeting • November 21-23, 2024 • Las Vegas, NV

INTRODUCTION

- Chronic low back pain is a common condition often treated with thermal ablation via RFA, which destroys specific nerves to provide pain relief[1,2]
- Cryoneurolysis is an alternative treatment that applies cold temperatures to disrupt nerve conduction pathways via Wallerian degeneration and allows for nerve regrowth[3]; however, data for treatment of low back pain via cryoneurolysis are sparse

METHODS

STUDY DESIGN

- This single-center, randomized controlled trial (NCT06016127) enrolled participants with facet-mediated chronic low back pain and received institutional review board approval from Advarra, Inc (Pro00062787)
- Eligible participants either (1) underwent 2 positive diagnostic medial branch blocks with only local anesthetic (ie, no steroids) under fluoroscopic guidance that resulted in ≥50% relief of primary (index) pain for the period of time the local anesthetic was used, or (2) had a history of positive response to RFA treatment (≥6 months before enrollment)
- Participants received lumbar RFA or cryoneurolysis administered in the lumbar spine at the target medial branch nerves encompassing 1 level above and below the involved vertebral levels (ie, blocked levels were L4, L5, and L5 [dorsal ramus] to S1 [lateral branch])
 - All patients who received RFA also received a mixture of 3 mL 0.25% bupivacaine, 2 mL 1% lidocaine, and 1 mL 40 mg/mL triamcinolone, divided and injected at each level
 - Following treatment, participants had follow-up calls each day from Days 1 to 6 and on Days 7, 15, 60, 90, 120, 150, 210, 240, 270, 300, 330, and 360
 - Participants also had follow-up visits on Days 30 and 180

OUTCOMES

- The main outcomes of the study included:
 - Safety
 - Pain scores on numerical rating scale (NRS; scores range from 0 ["no pain"] to 10 ["worst possible pain"])
 - Functional disability status on Oswestry Disability Index (ODI; score range, 0-50; lower scores reflect milder disability)
 - Participant satisfaction with pain management (score range, 1 ["extremely dissatisfied"] to 5 ["extremely satisfied"])
 - Patient's global impression of change (PGIC)
- The original study period was 180 days; however, after obtaining 6-month data, a study extension was requested and granted for an additional 6 months of follow-up (ie, up to 1 year)
 - Participants were given the option to continue or discontinue prior to the additional 6 months of follow-up
- The least squares means (LSMs) for study outcomes were calculated after adjustment for sex, baseline NRS, and tobacco use
 - Data for Day 210 were not collected for most participants because the study extension request was under evaluation and these data are not included

RESULTS

PARTICIPANT CHARACTERISTICS

- Of 30 total participants, 15 received RFA and 15 received cryoneurolysis
 - Age, body mass index, low back pain duration, and baseline ODI scores were similarly distributed between groups (Table 1)
- Participants in each group had received spine injections within the last 12 months (cryoneurolysis, 15/15; RFA, 14/15)
 - Most injections occurred in the lumbar spine (cryoneurolysis, 14/15; RFA, 14/15) and were primarily facet and epidural injections (cryoneurolysis, 12/14 and 9/14, respectively; RFA, 10/14 and 4/14, respectively)
- Per inclusion criteria, all participants had a successful trial of at least 2 diagnostic nerve blocks (DNBs) preceding treatment
 - After the DNB, participants in the cryoneurolysis group had 87.3% relief and participants in the RFA group had 95.0% relief
- There were no differences in baseline characteristics or outcomes at 180 days between participants who remained in the study and those who dropped out

Table 1. Demographics and Baseline Characteristics

	RFA (n=15)	Cryoneurolysis (n=15)	Total (N=30)
Age, mean (SD), y	63.1 (12.7)	66.0 (17.1)	64.5 (14.9)
Sex, n (%)			
Male	7 (46.7)	9 (60.0)	16 (53.3)
Female	8 (53.3)	6 (40.0)	14 (46.7)
BMI, mean (SD), kg/m^2	28.1 (5.0)	26.5 (6.4)	27.3 (5.7)
White race, n (%)	15 (100.0)	15 (100.0)	30 (100.0)
Non-Hispanic or Latino, n (%)	15 (100.0)	15 (100.0)	15 (100.0)
Duration of low back pain, mean (SD), y[a]	19.6 (16.2)	24.9 (19.7)	22.7 (18.2)
Average pain score over 24 hours on NRS, mean (SD)	7.1 (1.6)	6.5 (1.9)	6.8 (1.8)
ODI score, mean (SD)	18.7 (5.9)	18.5 (7.1)	18.6 (6.4)
Any spine injections, n (%)	14 (93.3)	15 (100.0)	29 (96.7)

[a]RFA, n=10; cryoneurolysis, n=14; total, n=24. BMI, body mass index; NRS, numerical rating scale; ODI, Oswestry Disability Index; RFA, radiofrequency ablation; SD, standard deviation.

PAIN OUTCOMES

- After Day 7, LSM NRS pain scores were numerically higher with RFA versus cryoneurolysis (Figure 1)
 - At Day 180, cryoneurolysis was associated with a significant decrease in NRS pain scores versus RFA (LSM [95% confidence interval (CI)], 3.1 [2.1-4.2] vs 5.4 [4.3-6.5]; *P*=0.01)
 - Cryoneurolysis was also associated with a significant decrease in NRS pain scores versus RFA at Day 360 (LSM [95% CI], 3.0 [1.4-4.7] vs 6.1 [4.5-7.7]; *P*=0.01).

Figure 1. Adjusted least squares mean NRS scores through 360 days.

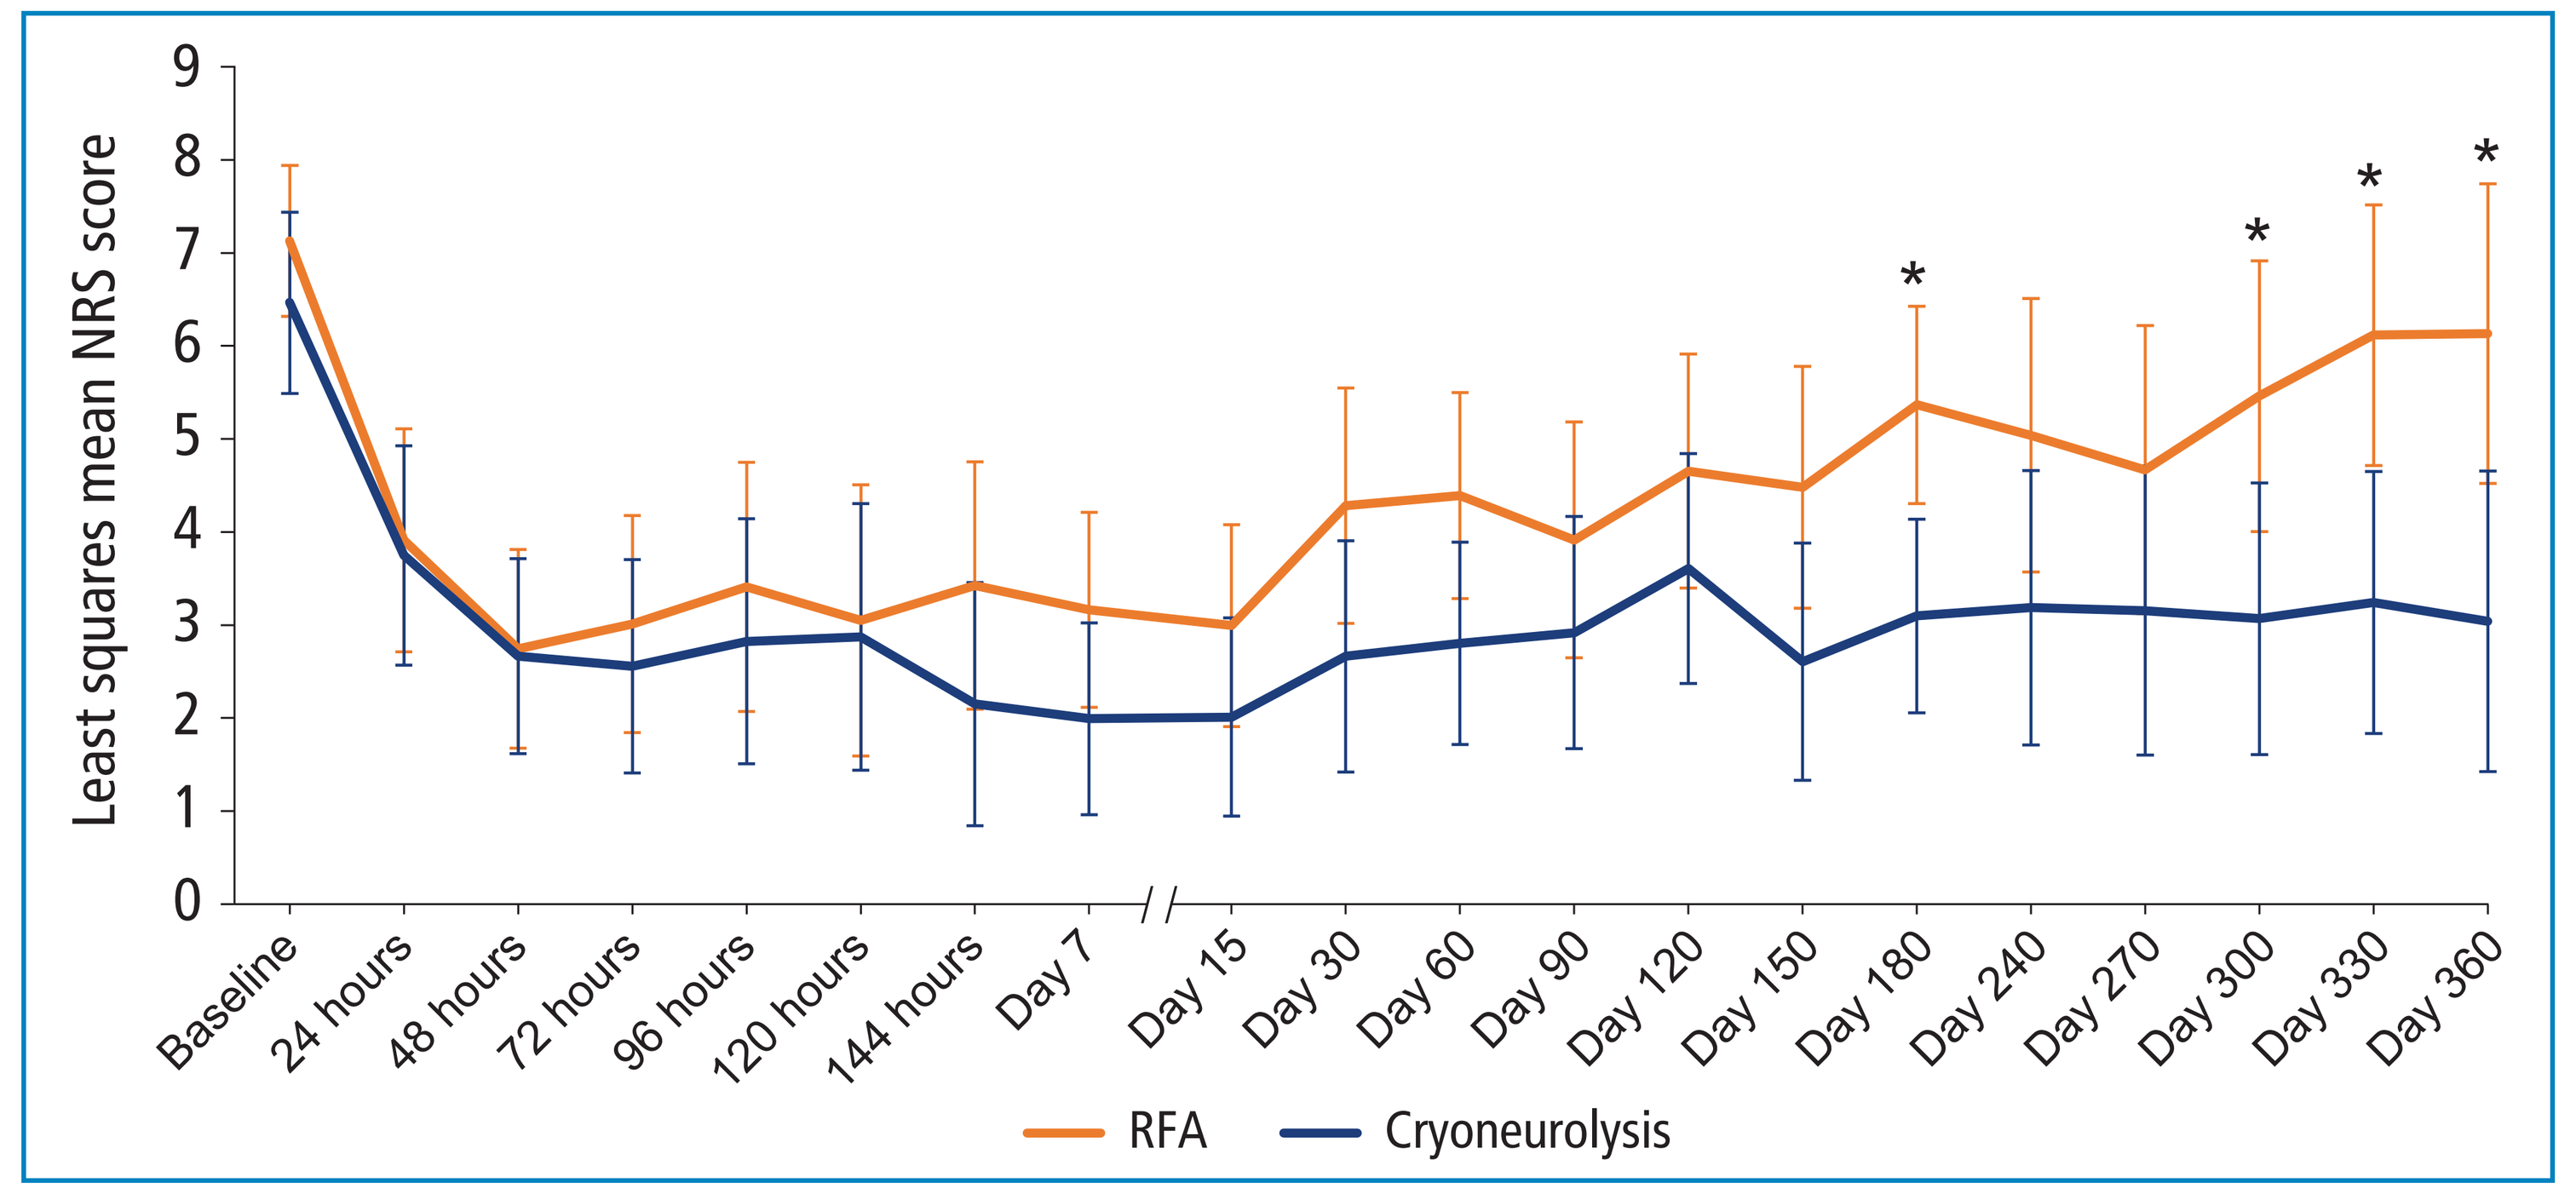

Least squares mean NRS score
0
1
2
3
4
5
6
7
8
9
Baseline
24 hours
48 hours
72 hours
96 hours
120 hours
144 hours
Day 7
Day 15
Day 30
Day 60
Day 90
Day 120
Day 150
Day 180
Day 240
Day 270
Day 300
Day 330
Day 360
*
RFA
Cryoneurolysis

Data for Day 210 excluded because an ongoing request to extend the study was under evaluation. Baseline least squares mean is not adjusted for covariates. Error bars are the 95% confidence interval. *$P<0.05$. NRS, numerical rating scale; RFA, radiofrequency ablation.

DISABILITY OUTCOMES

- At Day 360, ODI scores were significantly lower with cryoneurolysis versus RFA (LSM [95% CI], 10.2 [6.0-14.3] vs 20.6 [16.5-24.7]; *P*=0.002) (Figure 2)
 - At Day 180, ODI scores were numerically lower with cryoneurolysis versus RFA (LSM [95% CI, 13.3 [8.9-17.8] vs 18.1 [13.6-22.6]; *P*=0.15)
 - The mean percent decrease in ODI score from baseline was greater with cryoneurolysis compared with RFA at Day 360 (Figure 3)

Figure 2. Adjusted least squares mean ODI scores through 360 days.

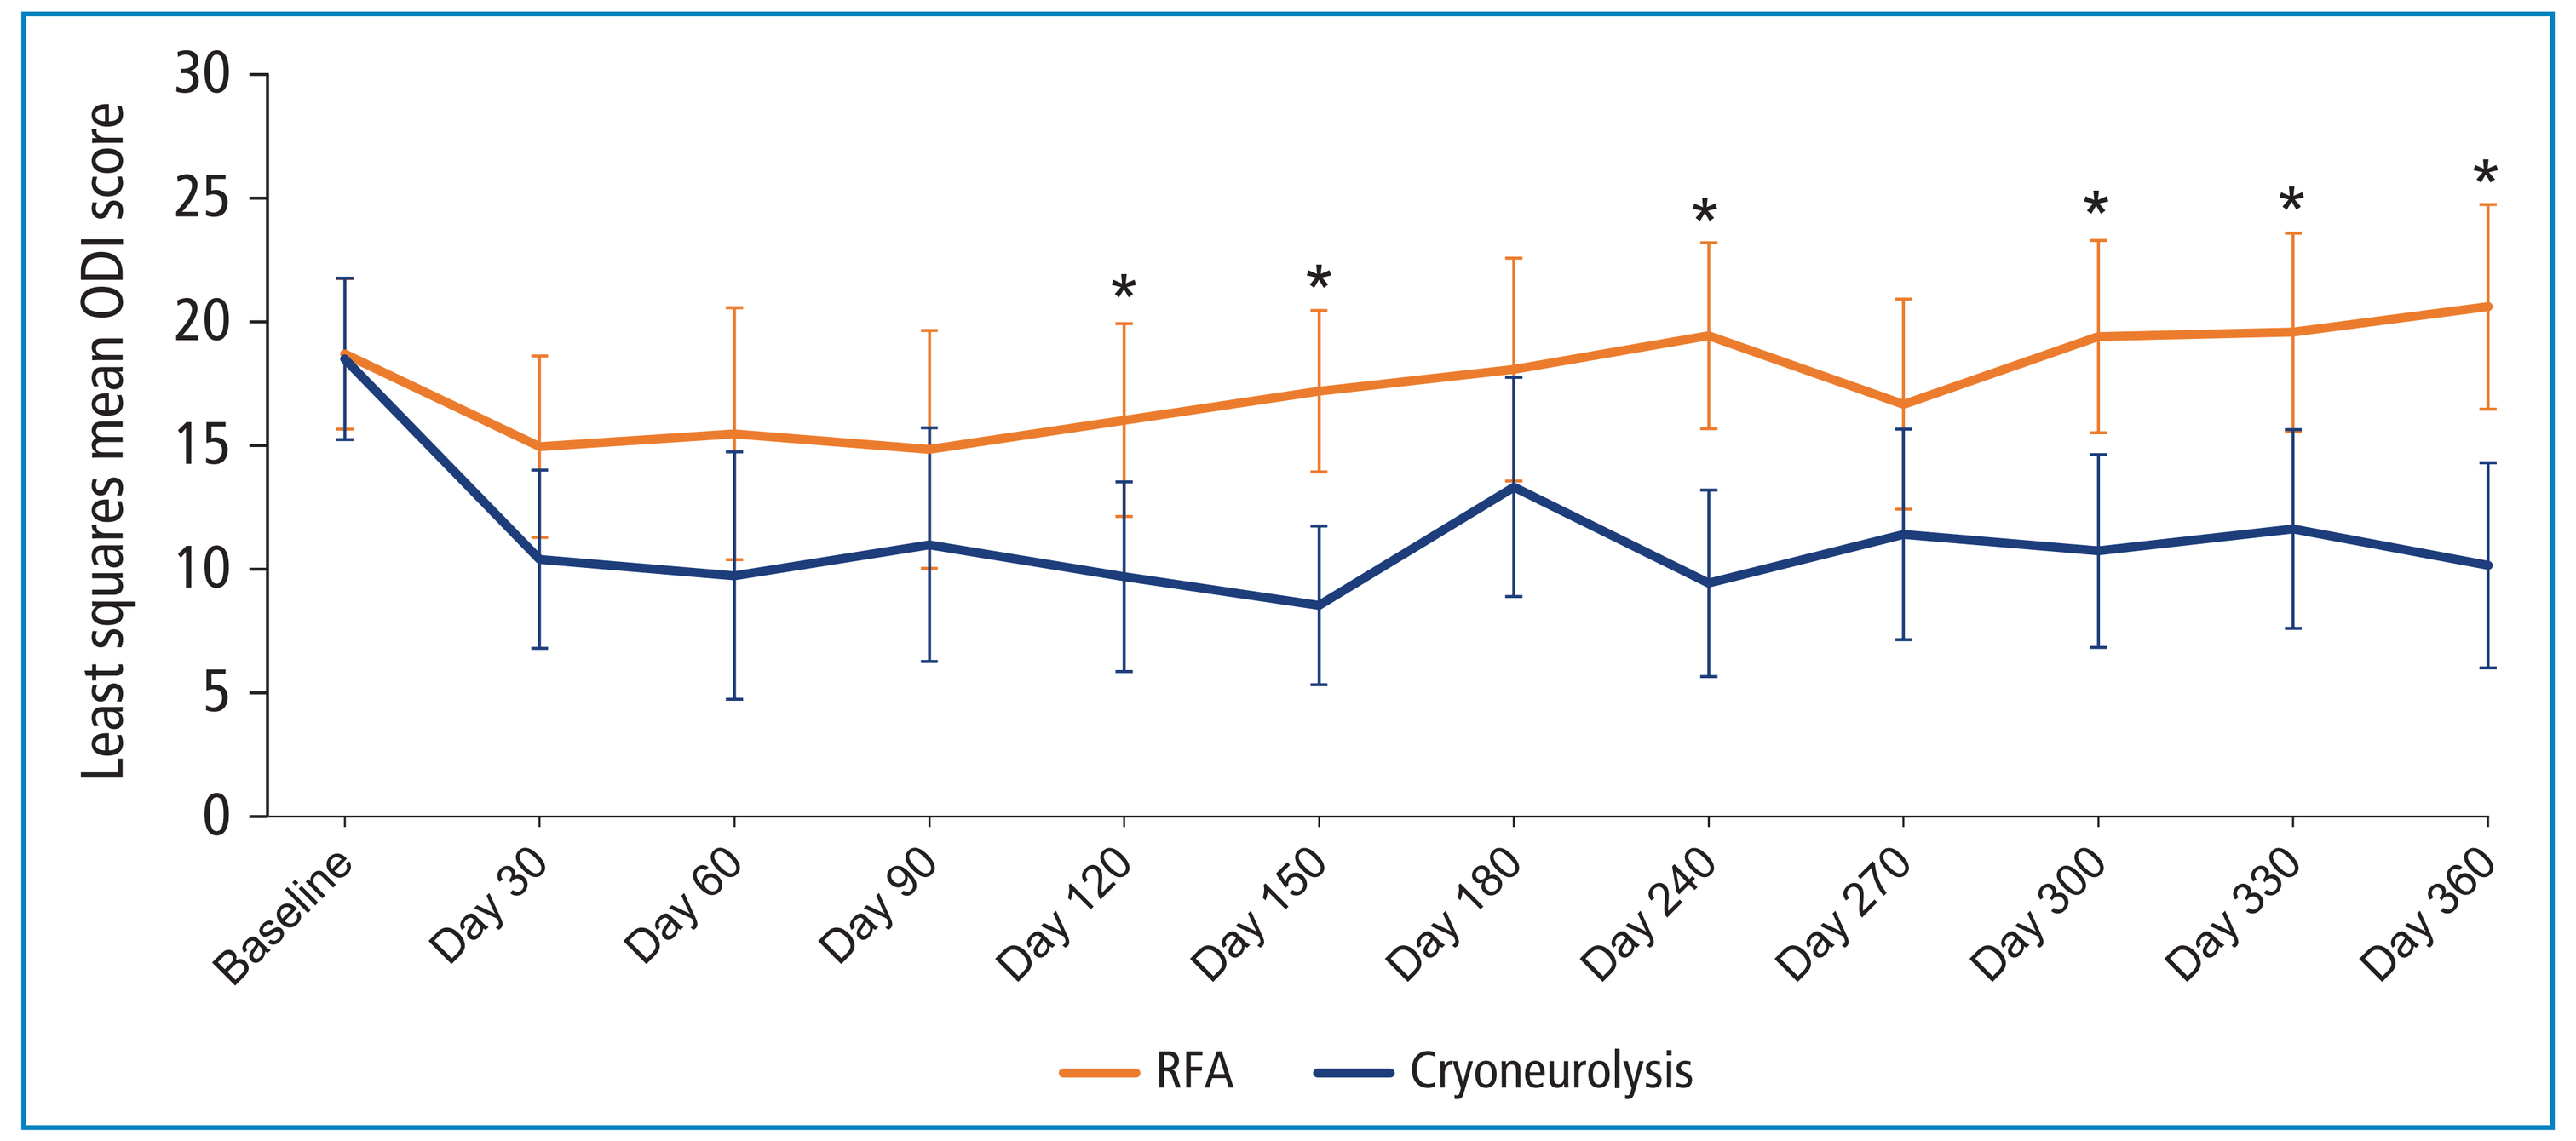

Least squares mean ODI score
0
5
10
15
20
25
30
Baseline
Day 30
Day 60
Day 90
Day 120
Day 150
Day 180
Day 240
Day 270
Day 300
Day 330
Day 360
*
RFA
Cryoneurolysis

Data for Day 210 excluded because an ongoing request to extend the study was under evaluation. Baseline least squares mean is not adjusted for covariates. Error bars are the 95% confidence interval. *$P<0.05$. ODI, Oswestry Disability Index; RFA, radiofrequency ablation.

Figure 3. Mean percent change in ODI from baseline through 360 days.

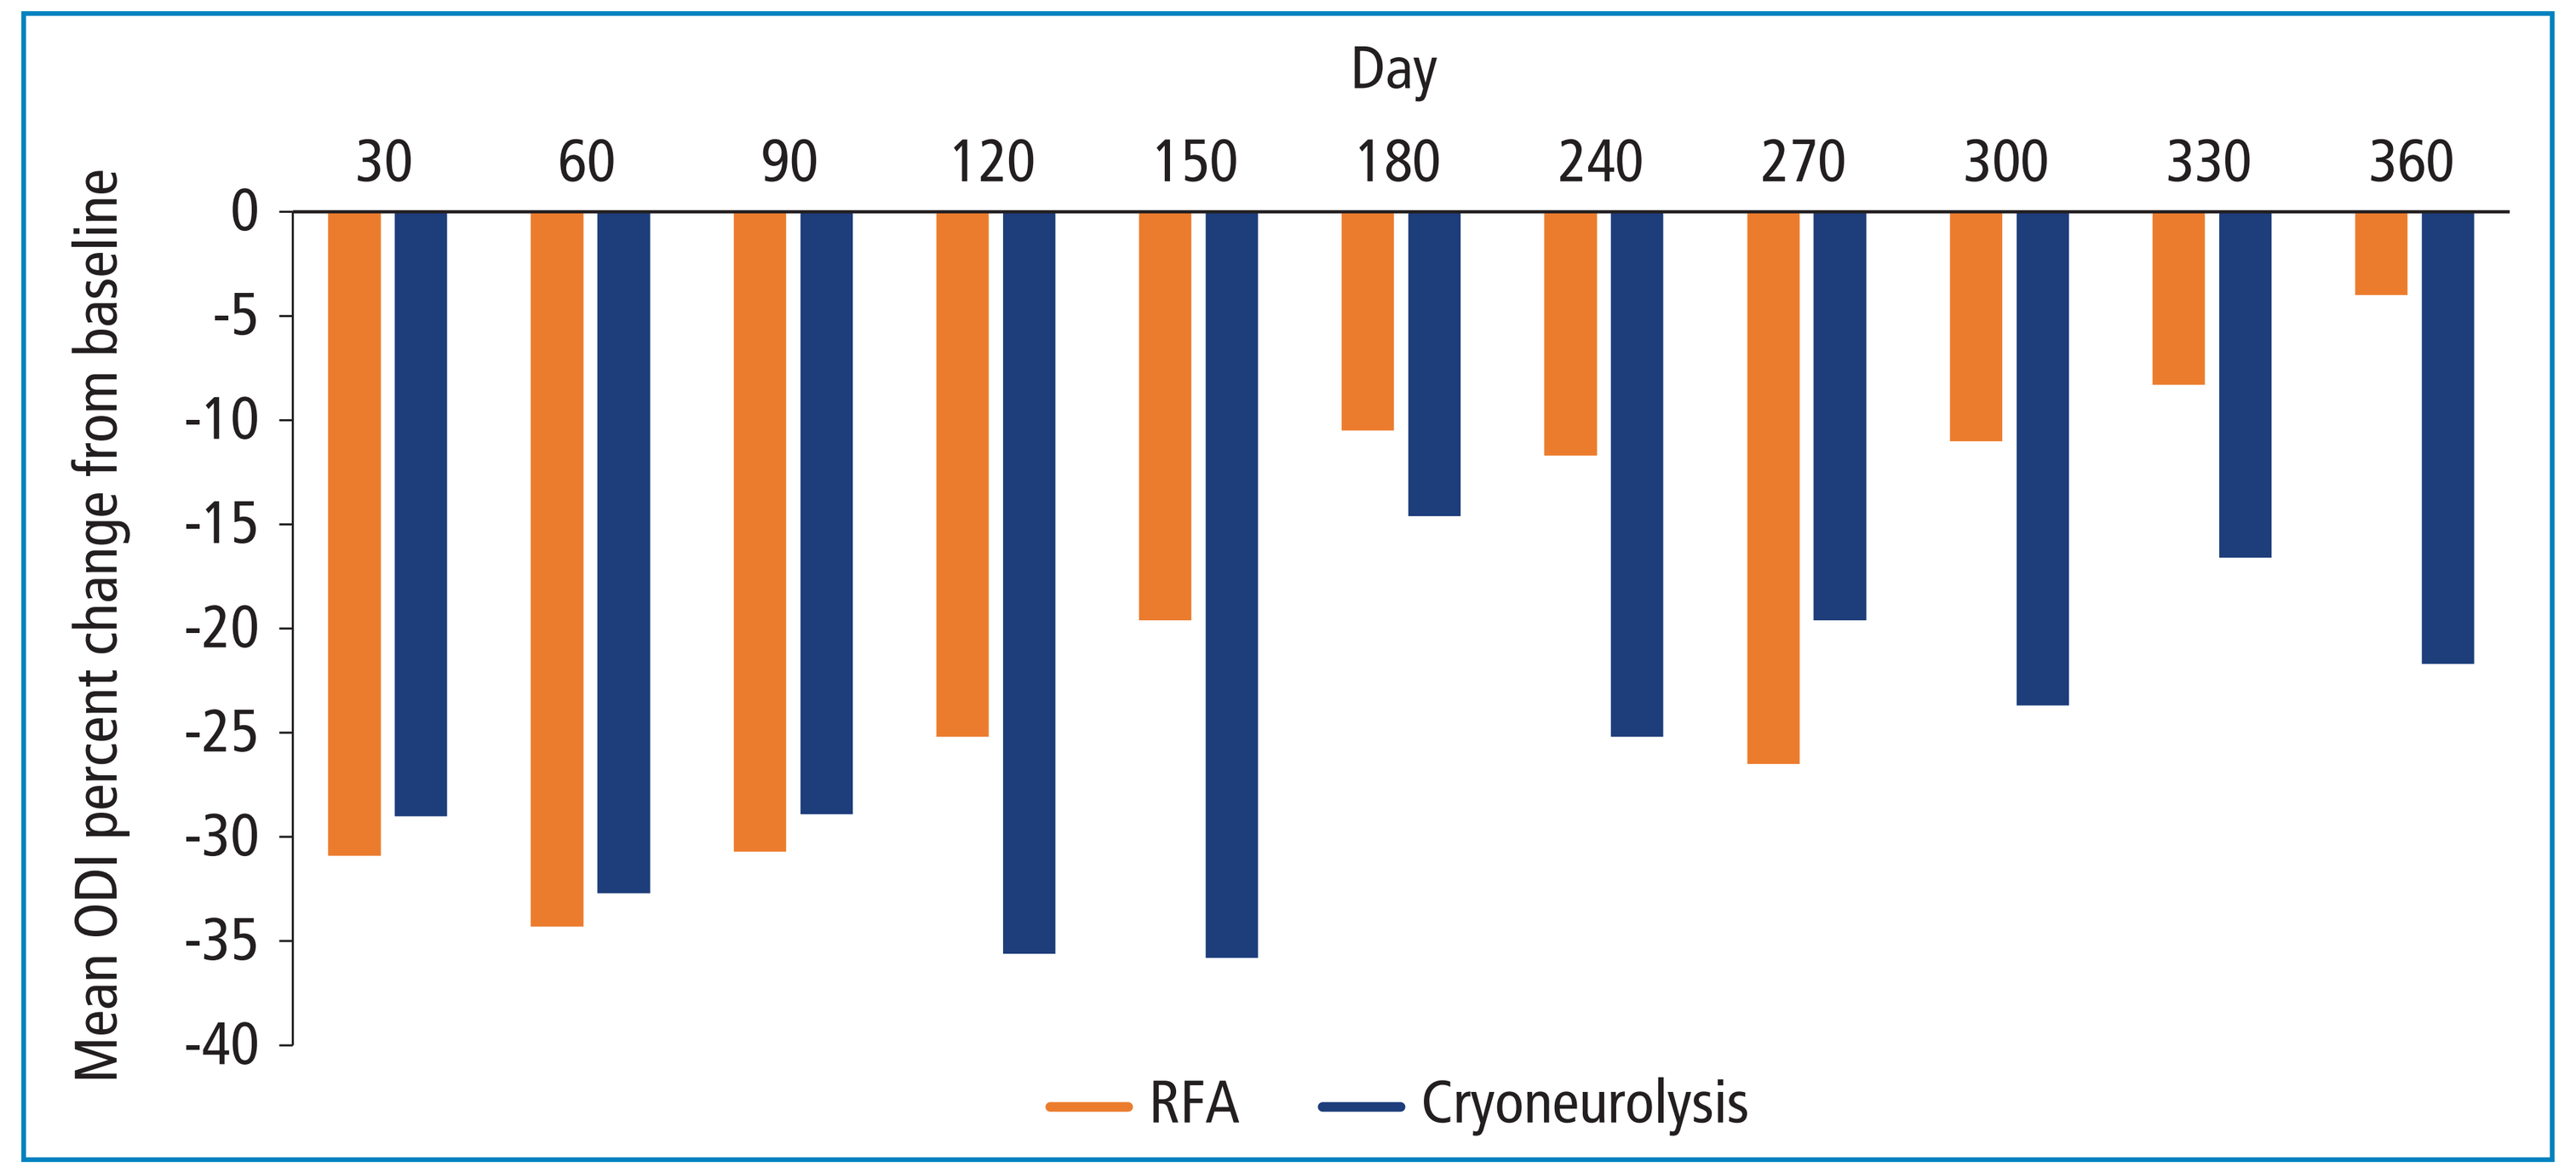

Day
30
60
90
120
150
180
240
270
300
330
360
Mean ODI percent change from baseline
0
-5
-10
-15
-20
-25
-30
-35
-40
RFA
Cryoneurolysis

ODI, Oswestry Disability Index; RFA, radiofrequency ablation.

- More participants receiving cryoneurolysis had "no disability" at Day 180, 270, and 360 than those receiving RFA (Table 2)

Table 2. Change in Disability Status Through 360 Days

Time	Treatment	n	No disability, n (%)	Mild, n (%)	Moderate, n (%)	Severe, n (%)	Completely disabled, n (%)
Baseline	RFA	15	0	3 (20.0)	10 (66.7)	2 (13.3)	0
	Cryoneurolysis	15	0	5 (33.3)	6 (40.0)	4 (26.7)	0
Day 60	RFA	15	3 (20.0)	8 (53.3)	2 (13.3)	1 (6.7)	1 (6.7)
	Cryoneurolysis	15	5 (33.3)	5 (33.3)	2 (13.3)	3 (20.0)	0
Day 180	RFA	15	0	7 (46.7)	5 (33.3)	3 (20.0)	0
	Cryoneurolysis	15	4 (26.7)	3 (20.0)	6 (40.0)	2 (13.3)	0
Day 270	RFA	12	1 (8.3)	5 (41.7)	5 (41.7)	1 (8.3)	0
	Cryoneurolysis	11	2 (18.2)	5 (45.5)	4 (36.4)	0	0
Day 360	RFA	12	0	5 (41.7)	5 (41.7)	2 (16.7)	0
	Cryoneurolysis	11	1 (9.1)	5 (45.5)	5 (45.5)	0	0

RFA, radiofrequency ablation.

PARTICIPANT IMPROVEMENT AND SATISFACTION

- Cryoneurolysis was associated with PGIC improvements versus RFA at Day 360 (LSM [95% CI], 1.7 [0.7-2.8] vs 4.4 [3.3-5.4]; *P*=0.002) (Figure 4)
 - Cryoneurolysis was also associated with numerical improvements at Day 180 (LSM [95% CI], 2.6 [1.6-3.7] vs 3.6 [2.6-4.7]; *P*=0.2)

Figure 4. Adjusted least squares mean for PGIC through 360 days.

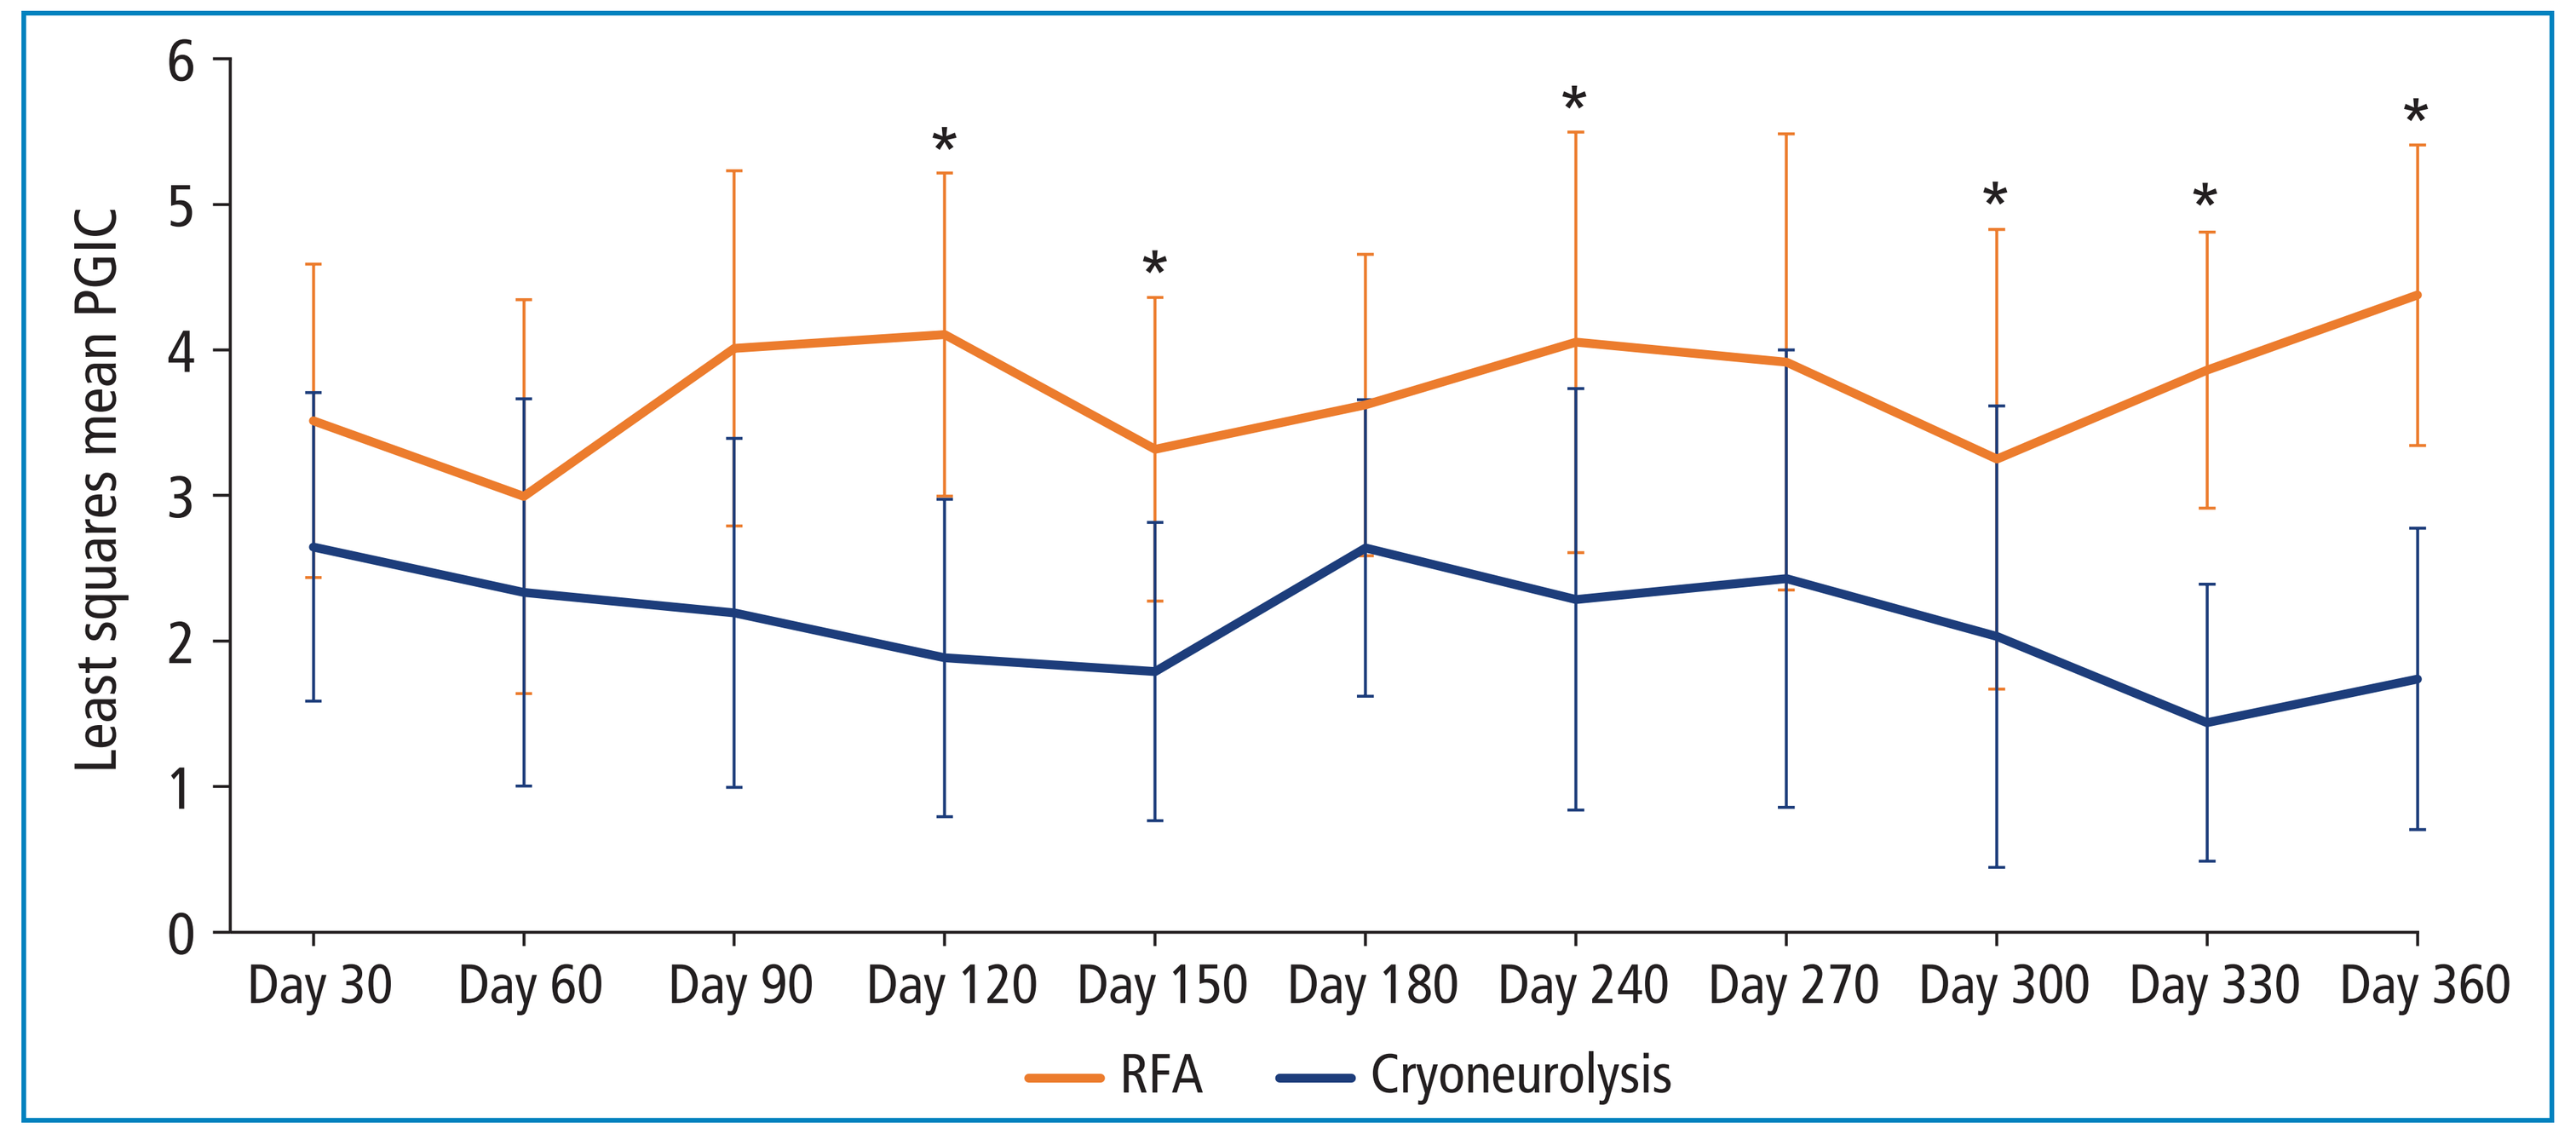

Least squares mean PGIC
0
1
2
3
4
5
6
Day 30
Day 60
Day 90
Day 120
Day 150
Day 180
Day 240
Day 270
Day 300
Day 330
Day 360
*
RFA
Cryoneurolysis

Error bars are the 95% confidence interval. *$P<0.05$. PGIC, patient's global impression of change; RFA, radiofrequency ablation.

- More participants were satisfied with pain management after cryoneurolysis versus RFA (90.9% vs 66.7%) at day 360 (Figure 5)

Figure 5. Participant satisfaction through 360 days.

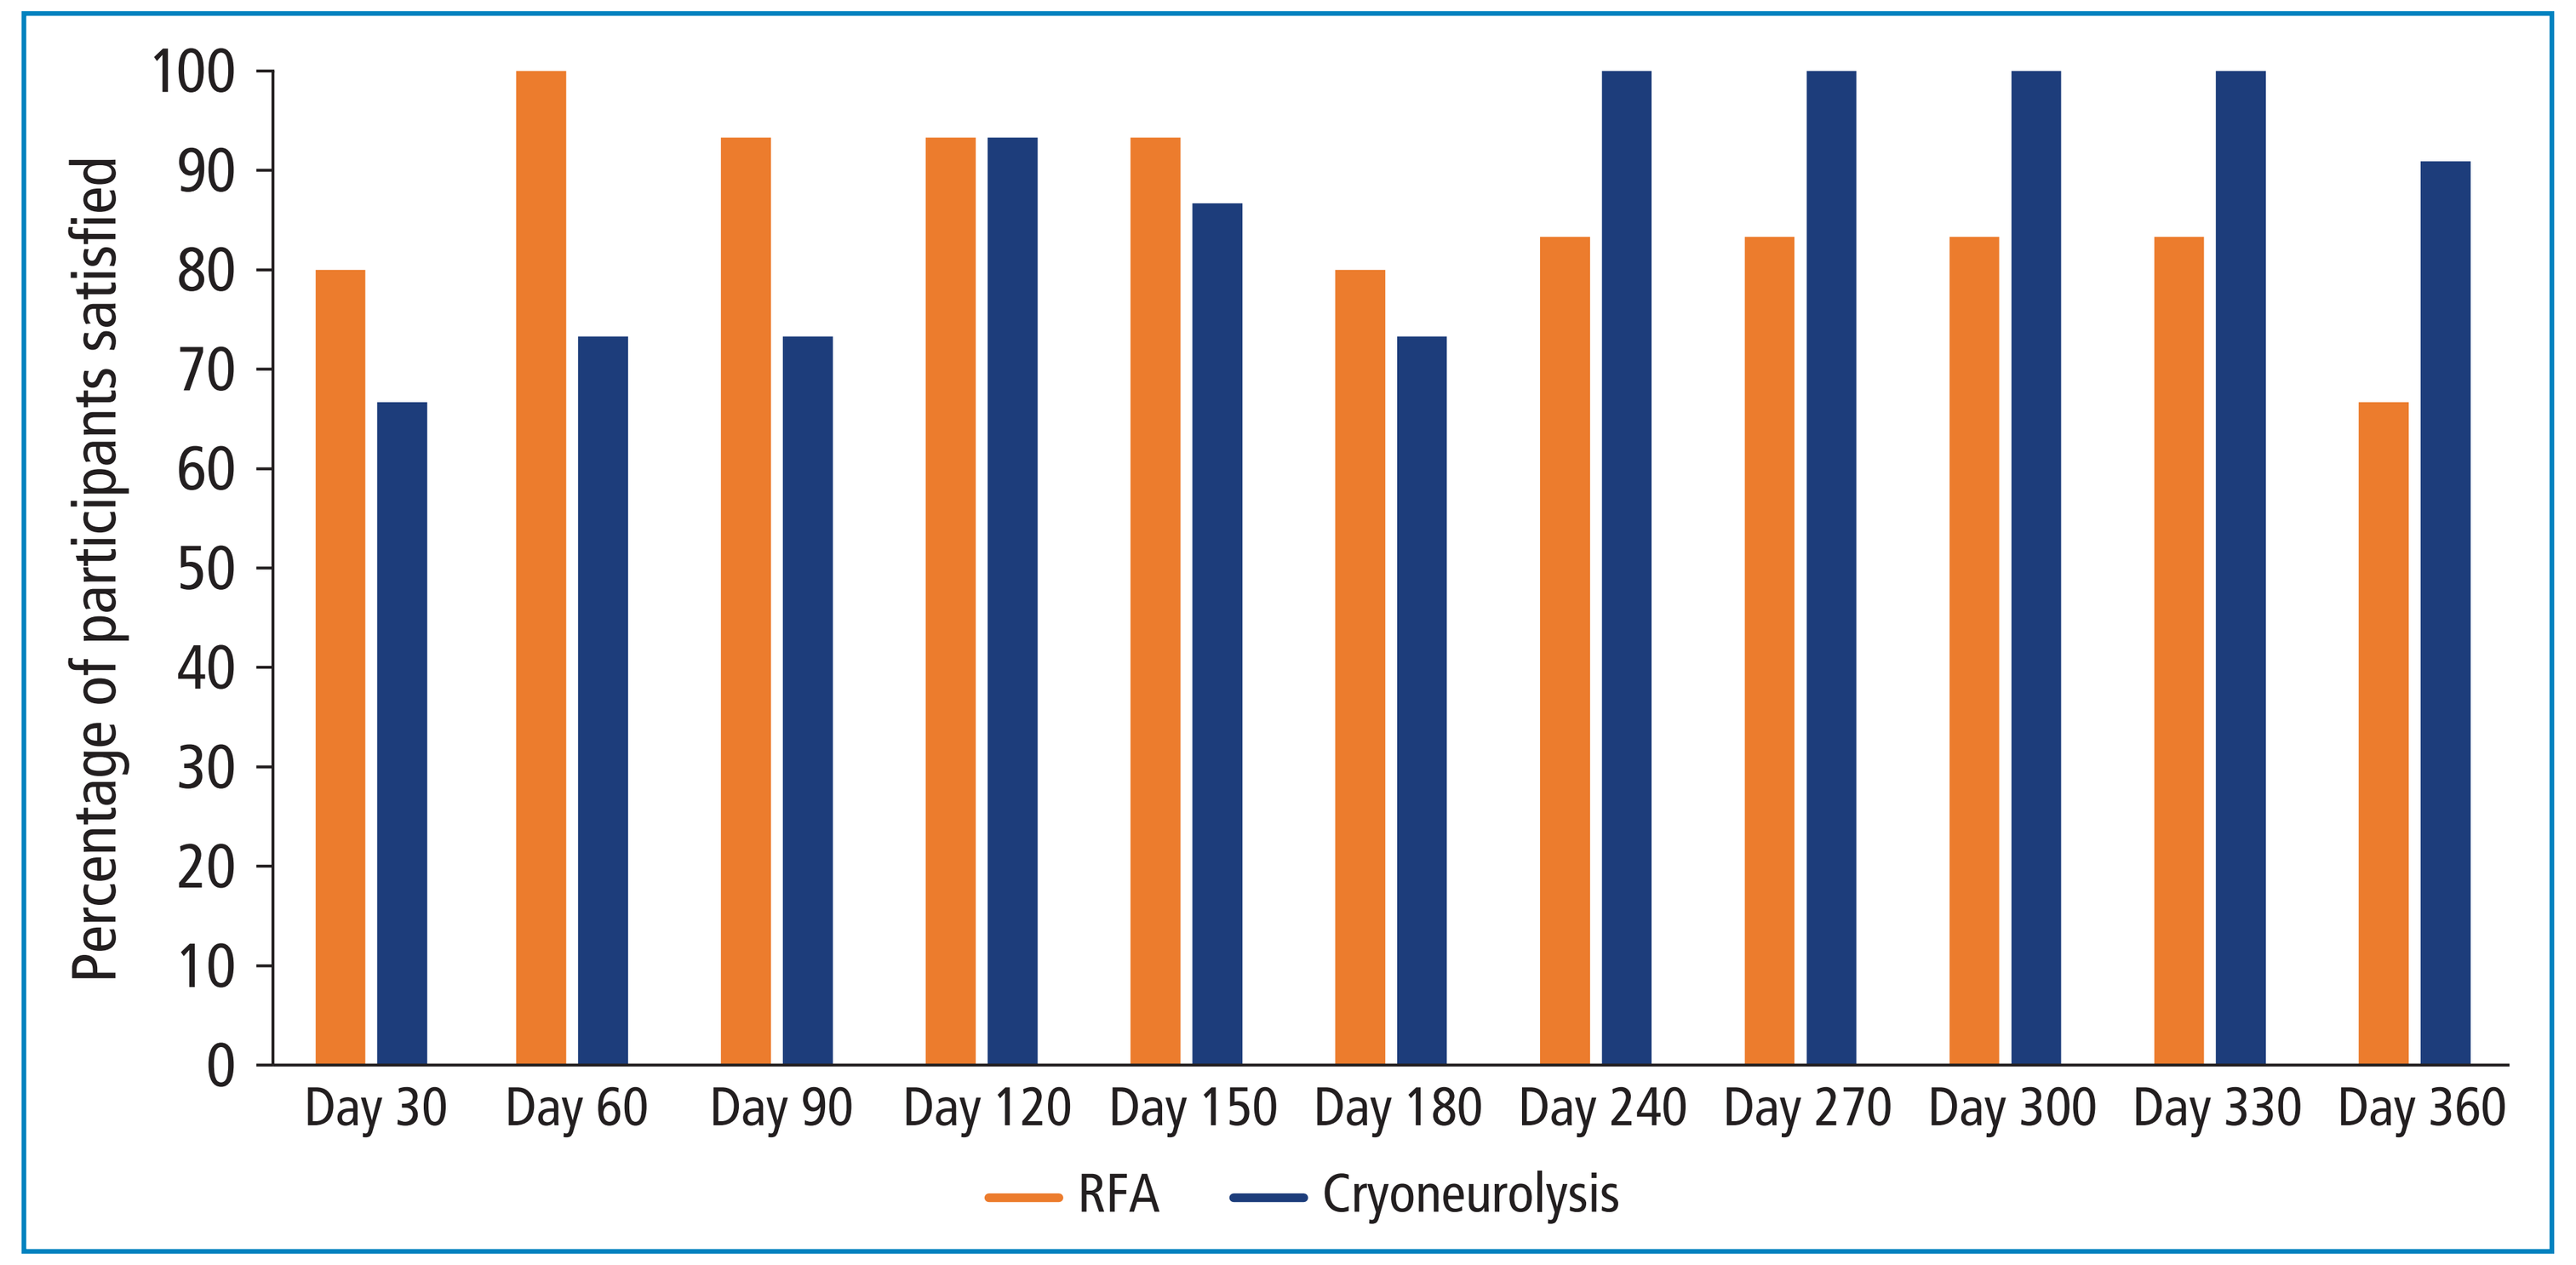

Percentage of participants satisfied
0
10
20
30
40
50
60
70
80
90
100
Day 30
Day 60
Day 90
Day 120
Day 150
Day 180
Day 240
Day 270
Day 300
Day 330
Day 360
RFA
Cryoneurolysis

RFA, radiofrequency ablation.

ADDITIONAL SPINAL INJECTIONS

- After Day 180, 5/11 (46%) and 9/12 (75%) participants receiving cryoneurolysis and RFA, respectively, required ≥1 additional spinal injection (facet, epidural, or spinal trigger point) in the cervical spine (RFA, n=3), thoracic spine (RFA, n=1), or lumbar spine (cryoneurolysis, n=7; RFA, n=8); some participants received ≥1 injection

SAFETY

- No serious adverse events were observed over the course of the study
- One mild adverse event was reported (compression fracture in the cryoneurolysis group) and was considered unrelated to study treatment by the investigator

DISCUSSION

- Participants who received cryoneurolysis without a steroid injection for low back pain had significant improvements in pain, disability, and overall impression of treatment through 1 year compared with participants who received RFA with a concomitant steroid injection
 - Additionally, participants who received cryoneurolysis were more satisfied with their pain management than those who received RFA with a concomitant steroid injection
- More participants who received RFA required additional spinal injections after Day 180 compared with those who received cryoneurolysis; cryoneurolysis treatment may reduce the need for additional treatments relative to RFA
- This pilot study revealed the potential for positive outcomes with cryoneurolysis relative to RFA with concomitant steroid injections; a large multicenter trial is warranted to confirm these results and further investigate the effects of cryoneurolysis on low back pain